Deep proteomic screening as a tool for systematic discovery and validation of degrader targets

Björn Schwalb, Uli Ohmayer, Martin Steger, Anastasia Helena Bednarz, Sophie Machata, Jutta Fritz and Henrik Daub

NEOsphere Biotechnologies GmbH, Fraunhoferstrasse 8, 82152 Martinsried, Germany

Targeted protein degradation (TPD) with molecular glue compounds is a breakthrough therapeutic modality to eliminate disease-relevant proteins previously considered undruggable. Despite this enormous potential, systematic discovery of novel molecular glues and their cellular degradation targets has so far been challenging. Here, we present a deep proteomic screening and validation platform to advance TPD drug discovery programs at all stages and create broad pipelines of novel, high-value targets. Deep proteomic screening is a data-independent acquisition (DIA) based MS technology to screen compound libraries of (potential) molecular glues against cellular proteomes at unprecedented throughput, coverage, and sensitivity. It identifies and quantifies more than 11,000 proteins per sample from cell lines treated with molecular glues, enabling comprehensive proteomics-based drug and drug target discovery.

Scalable proteomics compatible with drug discovery requirements

Cell treatment and automated sample preparation

- Automated and scalable **to efficiently and reproducibly screen entire degrader libraries of 10,000s of compounds at unparalleled turnaround times**
- Screening is performed in triplicates for every treatment condition
- Optimized to filter out noise and identify significant down-regulation of neosubstrate candidates

Single-shot LC-mass spectrometry

- Multiparameter optimization to maximize ion utilization and resolution in **data independent acquisition mass spectrometry** (DIA-PASEF[1])
- **Screening compatible**. Routine **coverage of up to 11,000 protein groups** (encoded by 10,500 genes) per sample
- **Data completeness of more than 99.5% at the protein level**

Data analysis & identification of novel targets

- Data processing using DIANN[3] optimized for **maximum coverage, precision and data completeness**
- Custom-made statistical analysis pipeline to **maximize the yield of putative neosubstrates**, through optimized filtering, normalization and batch correction procedures
- Reviewing of data and scoring of proteins to to **identify likely direct degrader targets**

Mechanistic validation of degrader targets

- Verification of E3 ligase and UPS dependency
- Detection of **degrader-induced E3 ligase binding by interactomics** (affinity enrichment-MS and cellular proximity labeling)
- **Ubiquitinomics** reliably quantifies **up to 50,000 ubiquitination sites** to demonstrate degrader-induced modifications[2]

Comparison of neosubstrate regulation for different degraders and cell lines

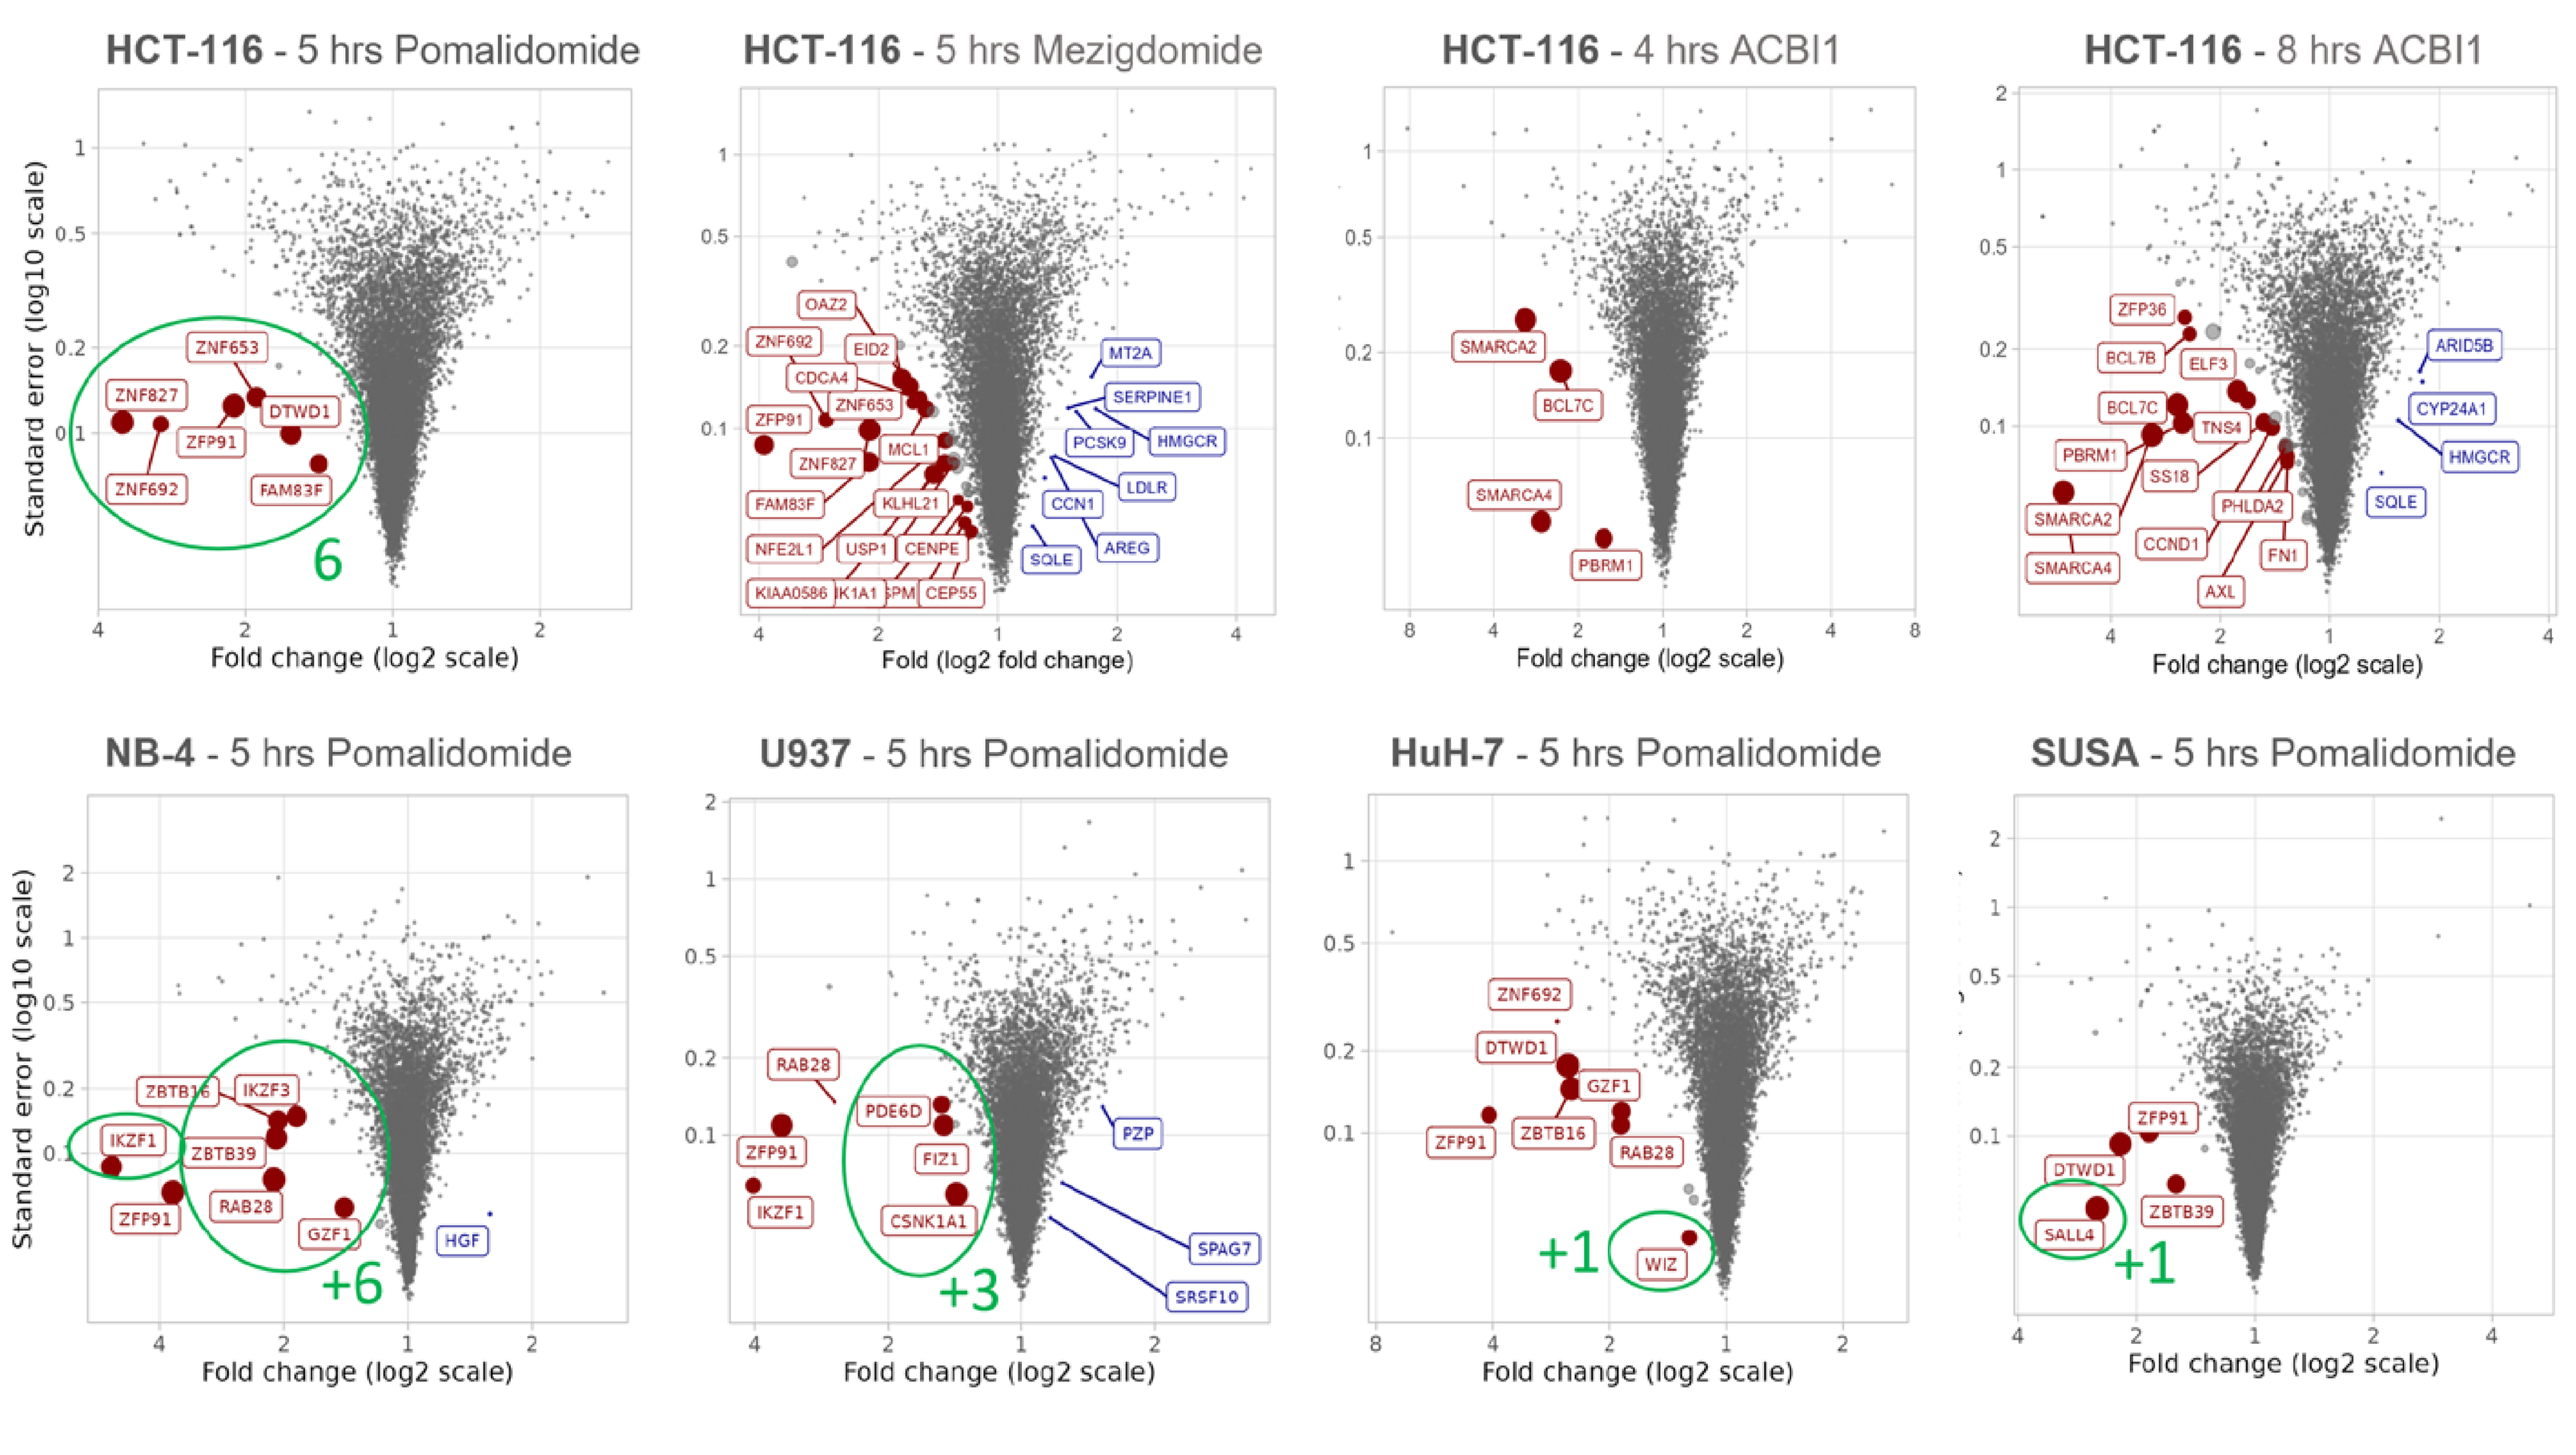
HCT-116 - 5 hrs Pomalidomide
HCT-116 - 5 hrs Mezigdomide
HCT-116 - 4 hrs ACBI1
HCT-116 - 8 hrs ACBI1
NB-4 - 5 hrs Pomalidomide
U937 - 5 hrs Pomalidomide
HuH-7 - 5 hrs Pomalidomide
SUSA - 5 hrs Pomalidomide
Standard error (log10 scale)
Fold change (log2 scale)
Fold (log2 fold change)
6
+6
+3
+1
+1

Comprehensive detection of compound-specific and cell line-specific degradation events

HCT-116 cells were treated with two IMiDs and one PROTAC as indicated (upper panel) and analyzed by single-shot MS analysis. The volcano plots illustrate significant up- (in blue) and downregulations (in red) induced by the different compounds. The x-axis depicts the fold change (log_2) of proteins in compound vs DMSO-treated cells and the y-axis the standard error. Known cellular targets such as zinc finger proteins were detected for pomalidomide and mezigdomide. SMARCA2 and SMARCA4 were downregulated upon 4 hrs treatment with the VHL based PROTAC ACBI1, together with two interacting BAF complex members, while secondary regulation was seen at a later 8 hrs time point. Additional cell lines were treated with pomalidomide for 5 hrs (lower panel) and compared to HCT-116. ZFP91 was significantly down-regulated in all cell lines, other neosubstrates exhibited varying degrees of regulation, reflecting cell type-specific expression (e.g., IKZF1 in U937 and NB-4, or SALL4 in SUSA) or different levels of IMiD responsiveness to commonly expressed neosubstrates. In total, 17 different neosubstrates were significantly down-regulated in at least one cell line.

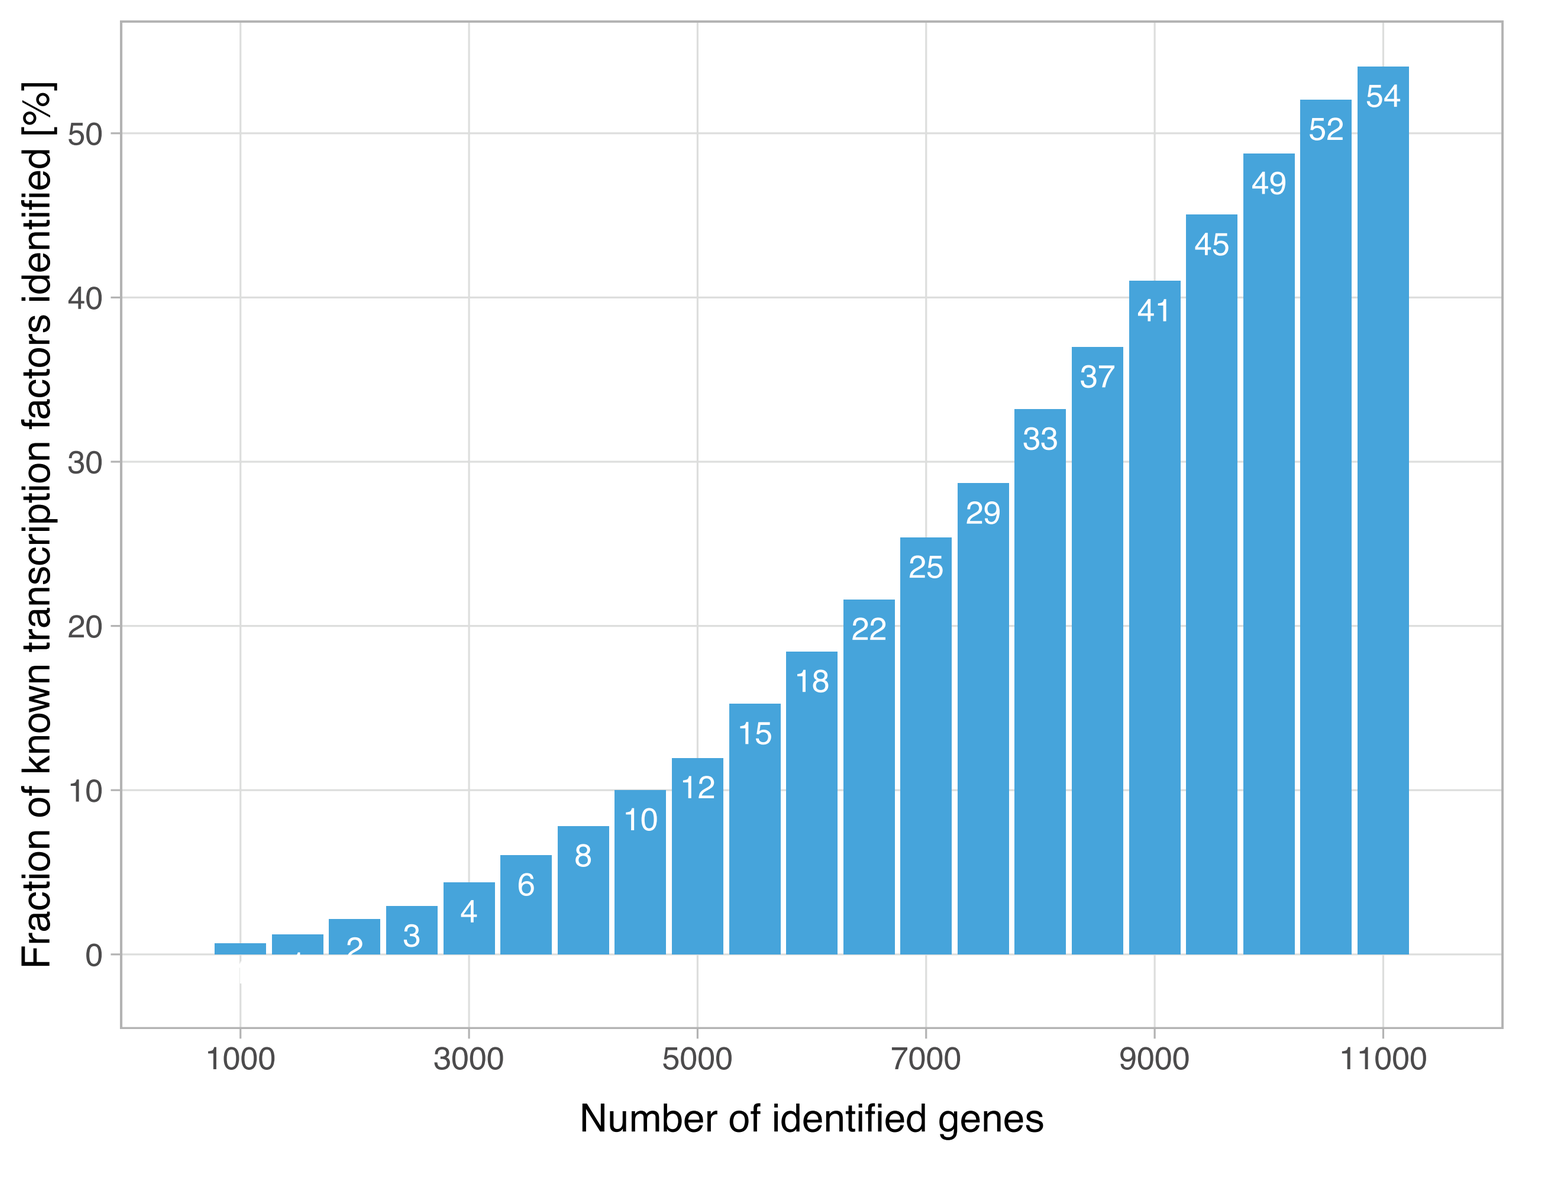
Fraction of known transcription factors identified [%]
Number of identified genes
1000
3000
5000
7000
9000
11000
2
3
4
6
8
10
12
15
18
22
25
29
33
37
41
45
49
52
54

Sensitive detection of low-abundance targets

A deep proteome coverage drastically increases the detection of low abundance proteins such as transcription factors. Among the ~11,000 proteins identified and quantified in one sample, more than half of all transcription factors encoded by the human genome can be detected. As transcription factors are overrepresented among low abundance proteins, twice as many are detected at a coverage of 11,000 compared to ~8,000 proteins, highlighting the importance of deep proteome coverage for the identification of degrader targets.

The effect of compound modification on cellular regulation

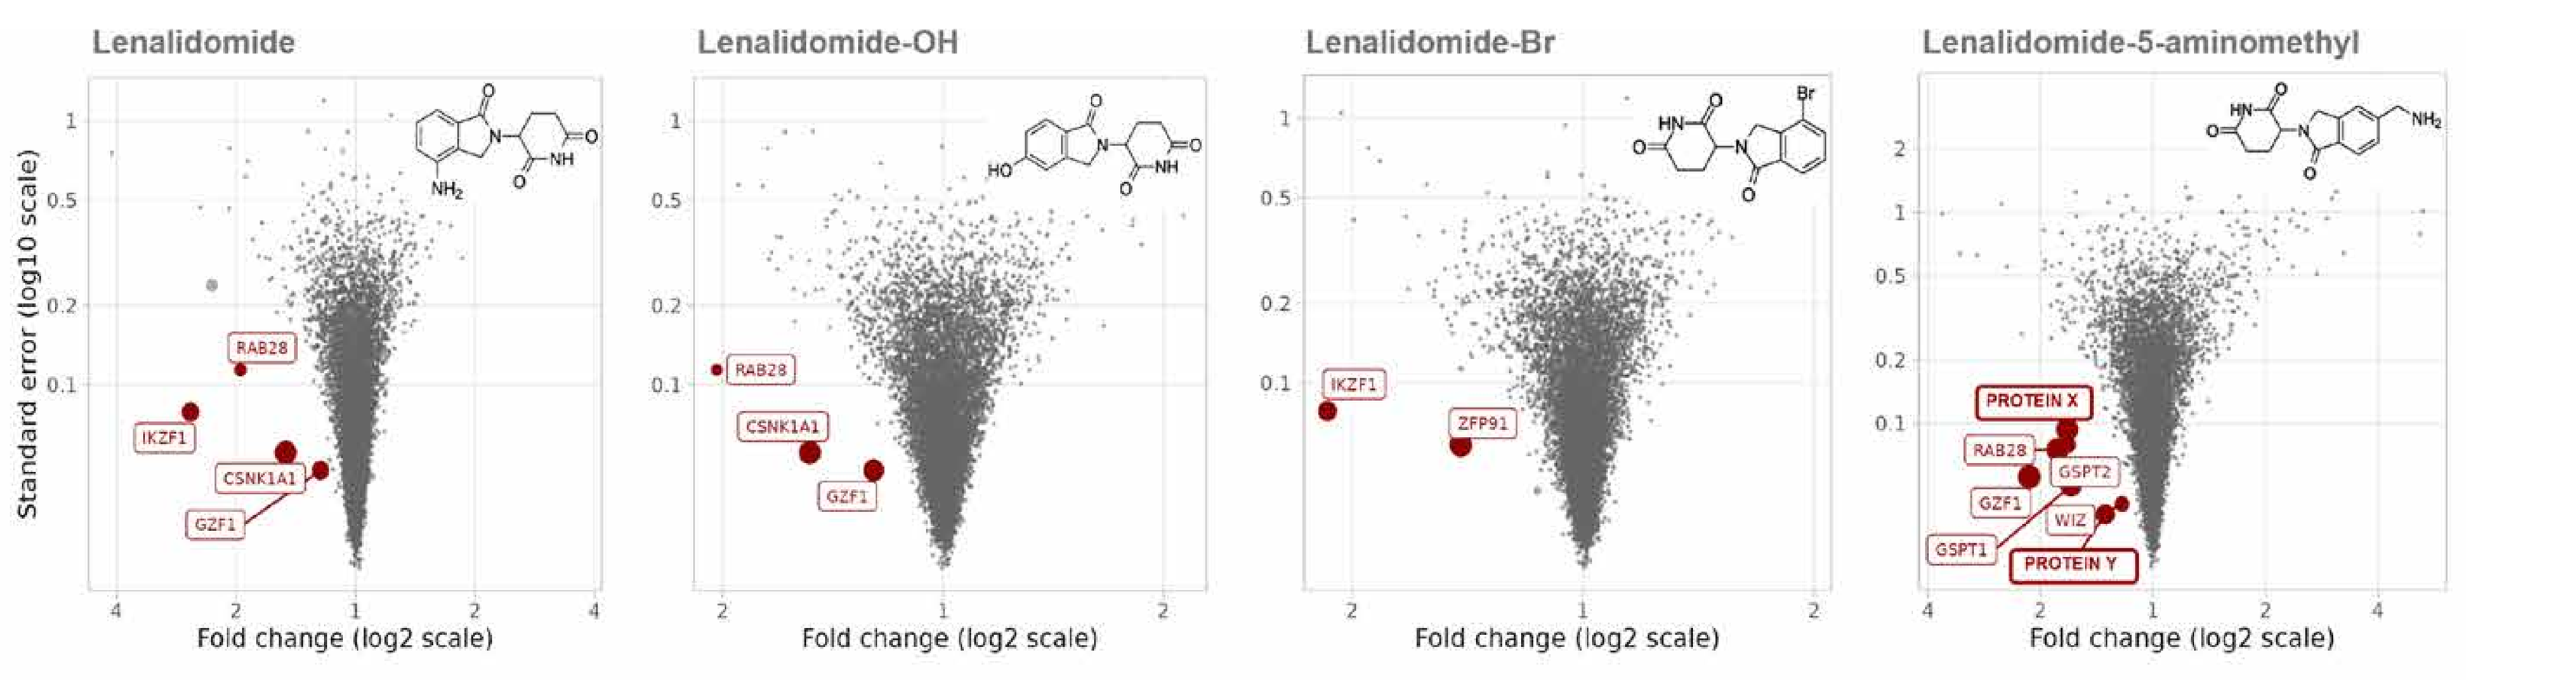
Lenalidomide
Lenalidomide-OH
Lenalidomide-Br
Lenalidomide-5-aminomethyl
Standard error (log10 scale)
Fold change (log2 scale)
RAB28
IKZF1
CSNK1A1
GZF1
ZFP91
PROTEIN X
GSPT2
GSPT1
WIZ
PROTEIN Y

Neosubstrate regulation by structurally related molecular glue compounds in NB-4 cells

NB-4 cells were treated with the indicated compounds for 5 hrs and analyzed by single-shot MS-analysis. Lenalidomide down-regulated four known neosubstrates, including IKZF1 and CSNK1A1. Subtle chemical changes had a profound effect on cellular selectivity, as shown by the absence of IKZF1 or CSNK1A1 degradation upon treatment with the derivatives lenalidomide-OH and lenalidomide-Br, respectively. Notably, lenalidomide-5-aminiomethyl significantly down-regulated additional proteins such as GSPT1/2 and two more proteins not yet described as neosubstrates in the scientific literature.

IMiD-induced neosubstrate binding and ternary complex formation

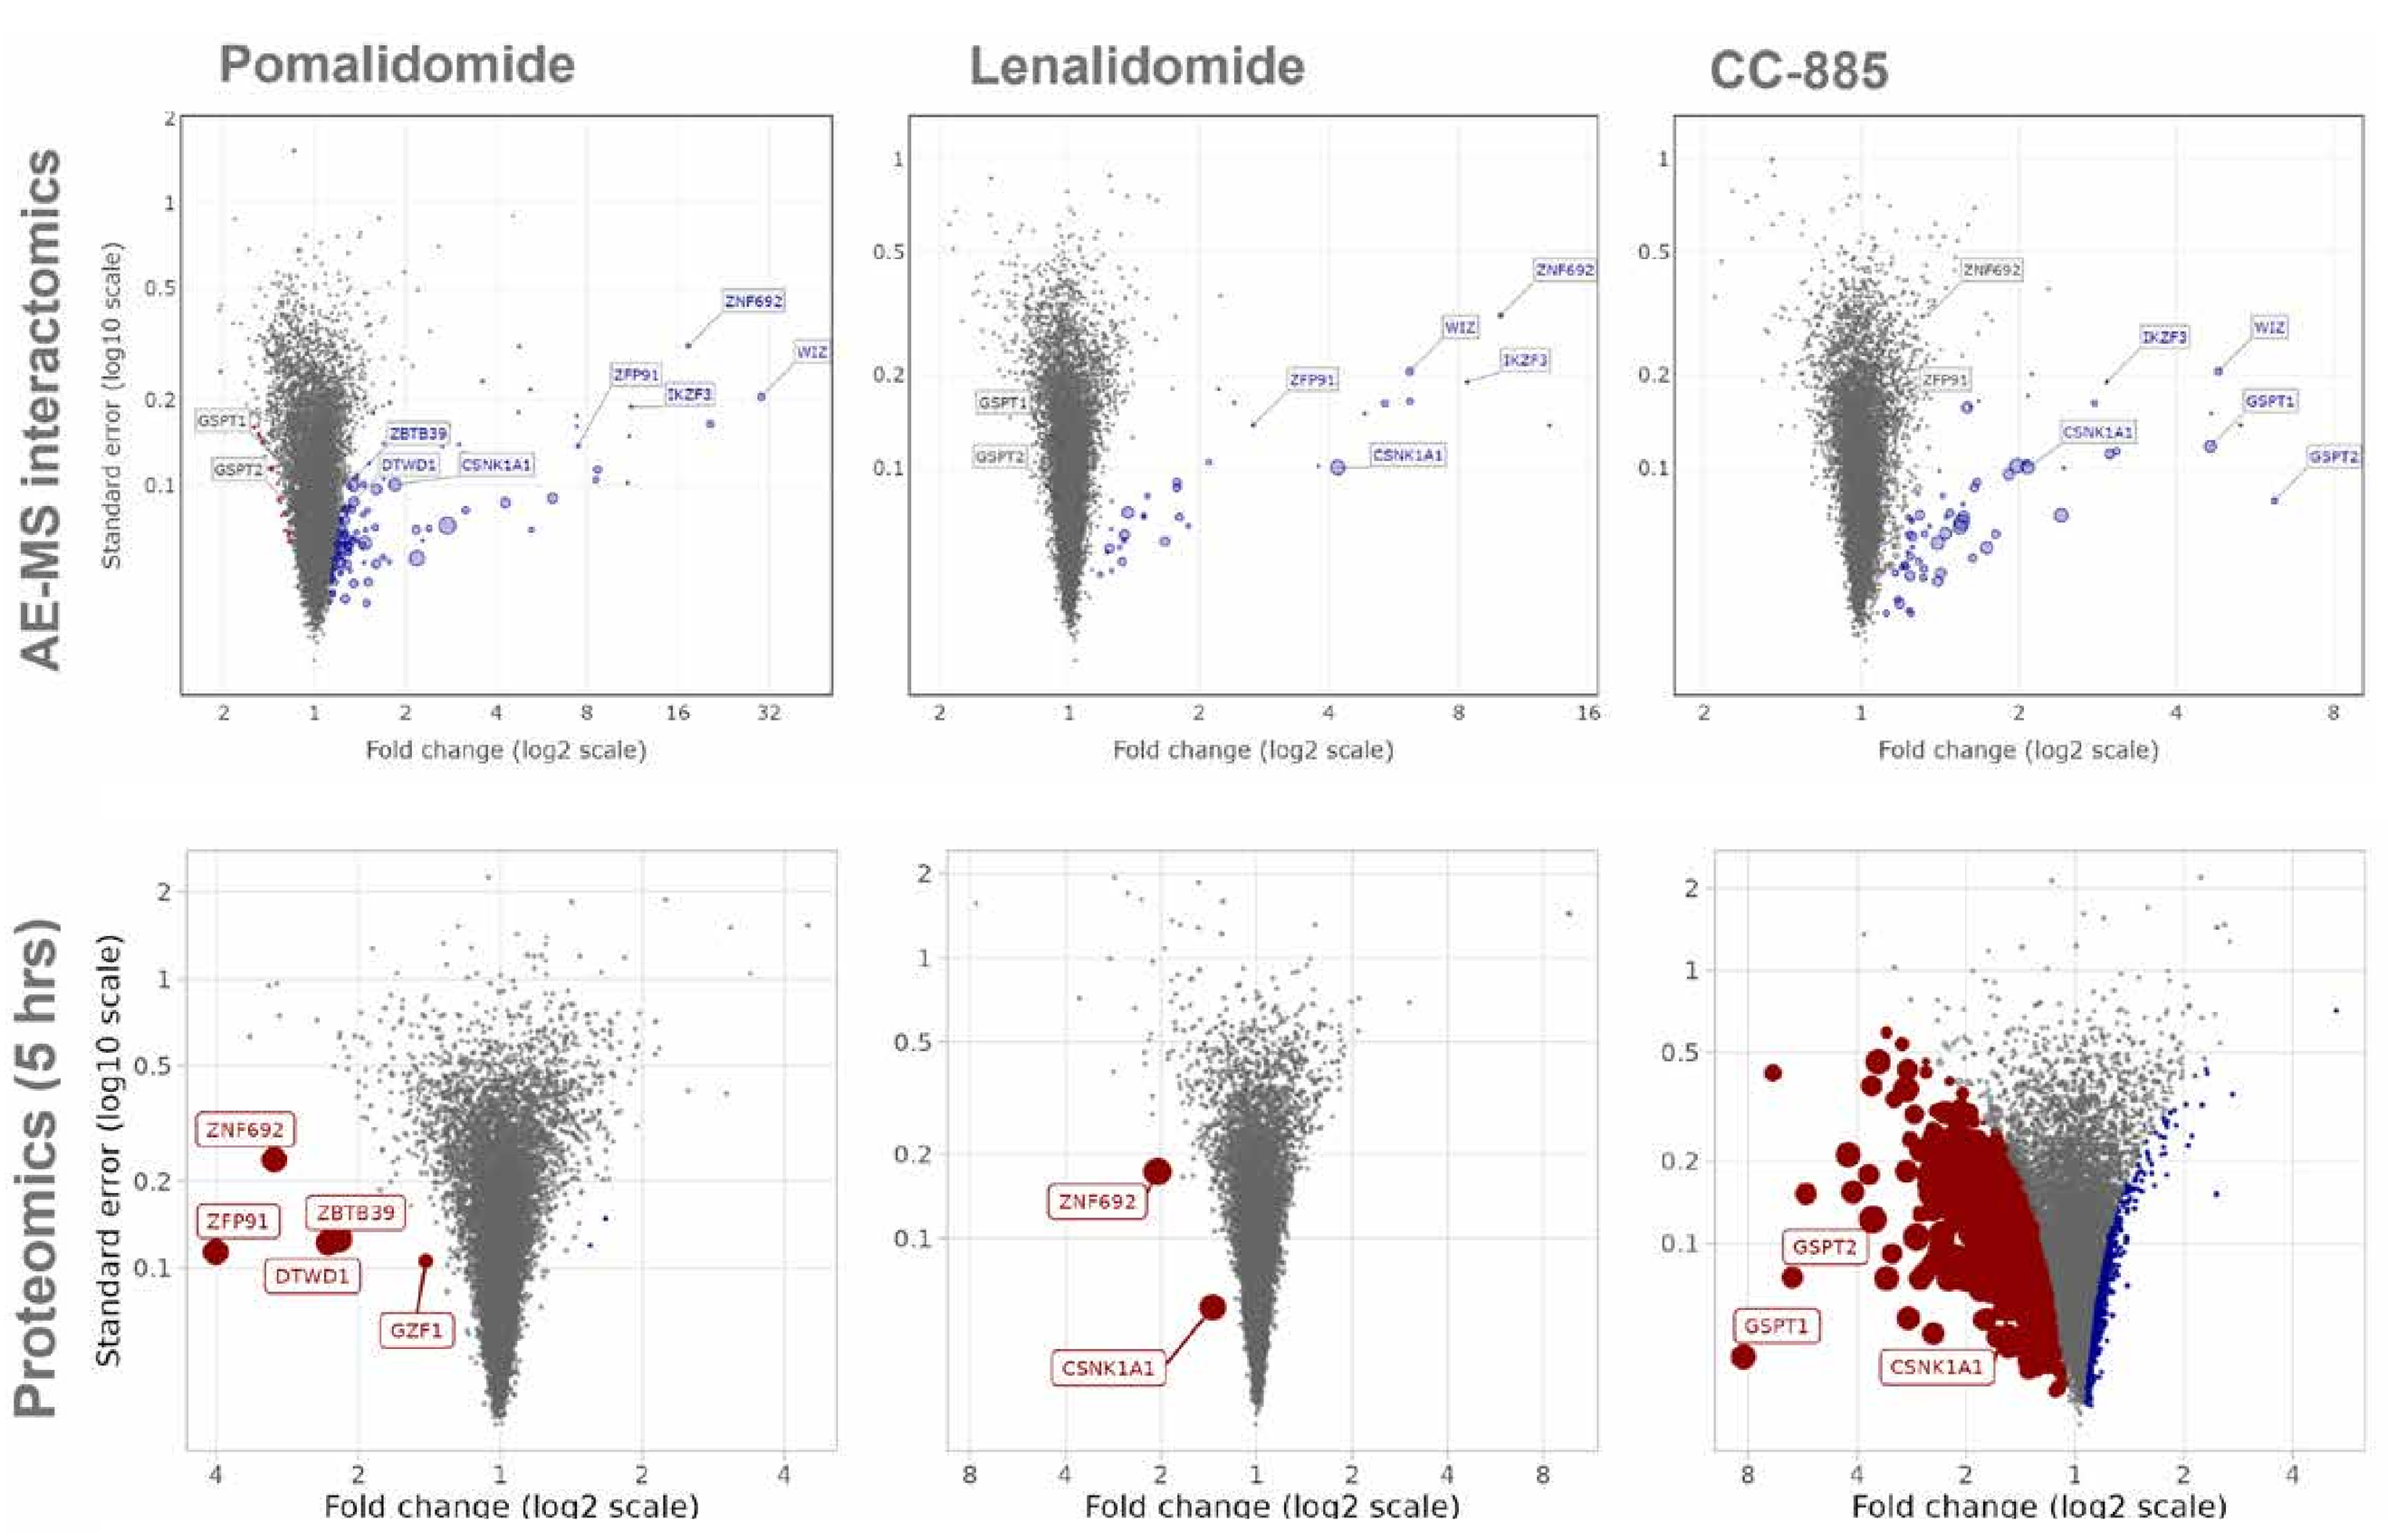
Pomalidomide
Lenalidomide
CC-885
AE-MS interactomics
Proteomics (5 hrs)
Standard error (log10 scale)
Fold change (log2 scale)
ZNF692
ZFP91
ZBTB39
DTWD1
GZF1
CSNK1A1
GSPT1
GSPT2
IKZF3
WIZ

Efficient interactomics workflow using affinity enrichment-MS and GFP-E3 ligase baits

Automated and optimized 96-well interactomics methods were established to perform affinity-enrichment mass spectrometry with GFP-cereblon expressing cell lysate. Upon addition of cereblon molecular glues such as pomalidomide, lenalidomide or CC-885, in vitro binding was induced for most neosubstrates down-regulated in cells (5 hrs proteomics, lower panel), such as ZFP91 for pomalidomide, casein kinase 1 alpha for lenalidomide, or GSPT1 and GSPT2 for CC-885. These and other significant compound-induced interactions are highlighted in blue in the interactomics experiments (upper panel). IMiD-dependent binding to GFP-CRBN was also seen for potential neosubstrates that were not degraded (WIZ) or not detected (IKZF3) in HEK293 cells, indicating high sensitivity and potential to discover new neosubstrate candidates.

Confirmation of neosubstrate ubiquitination events

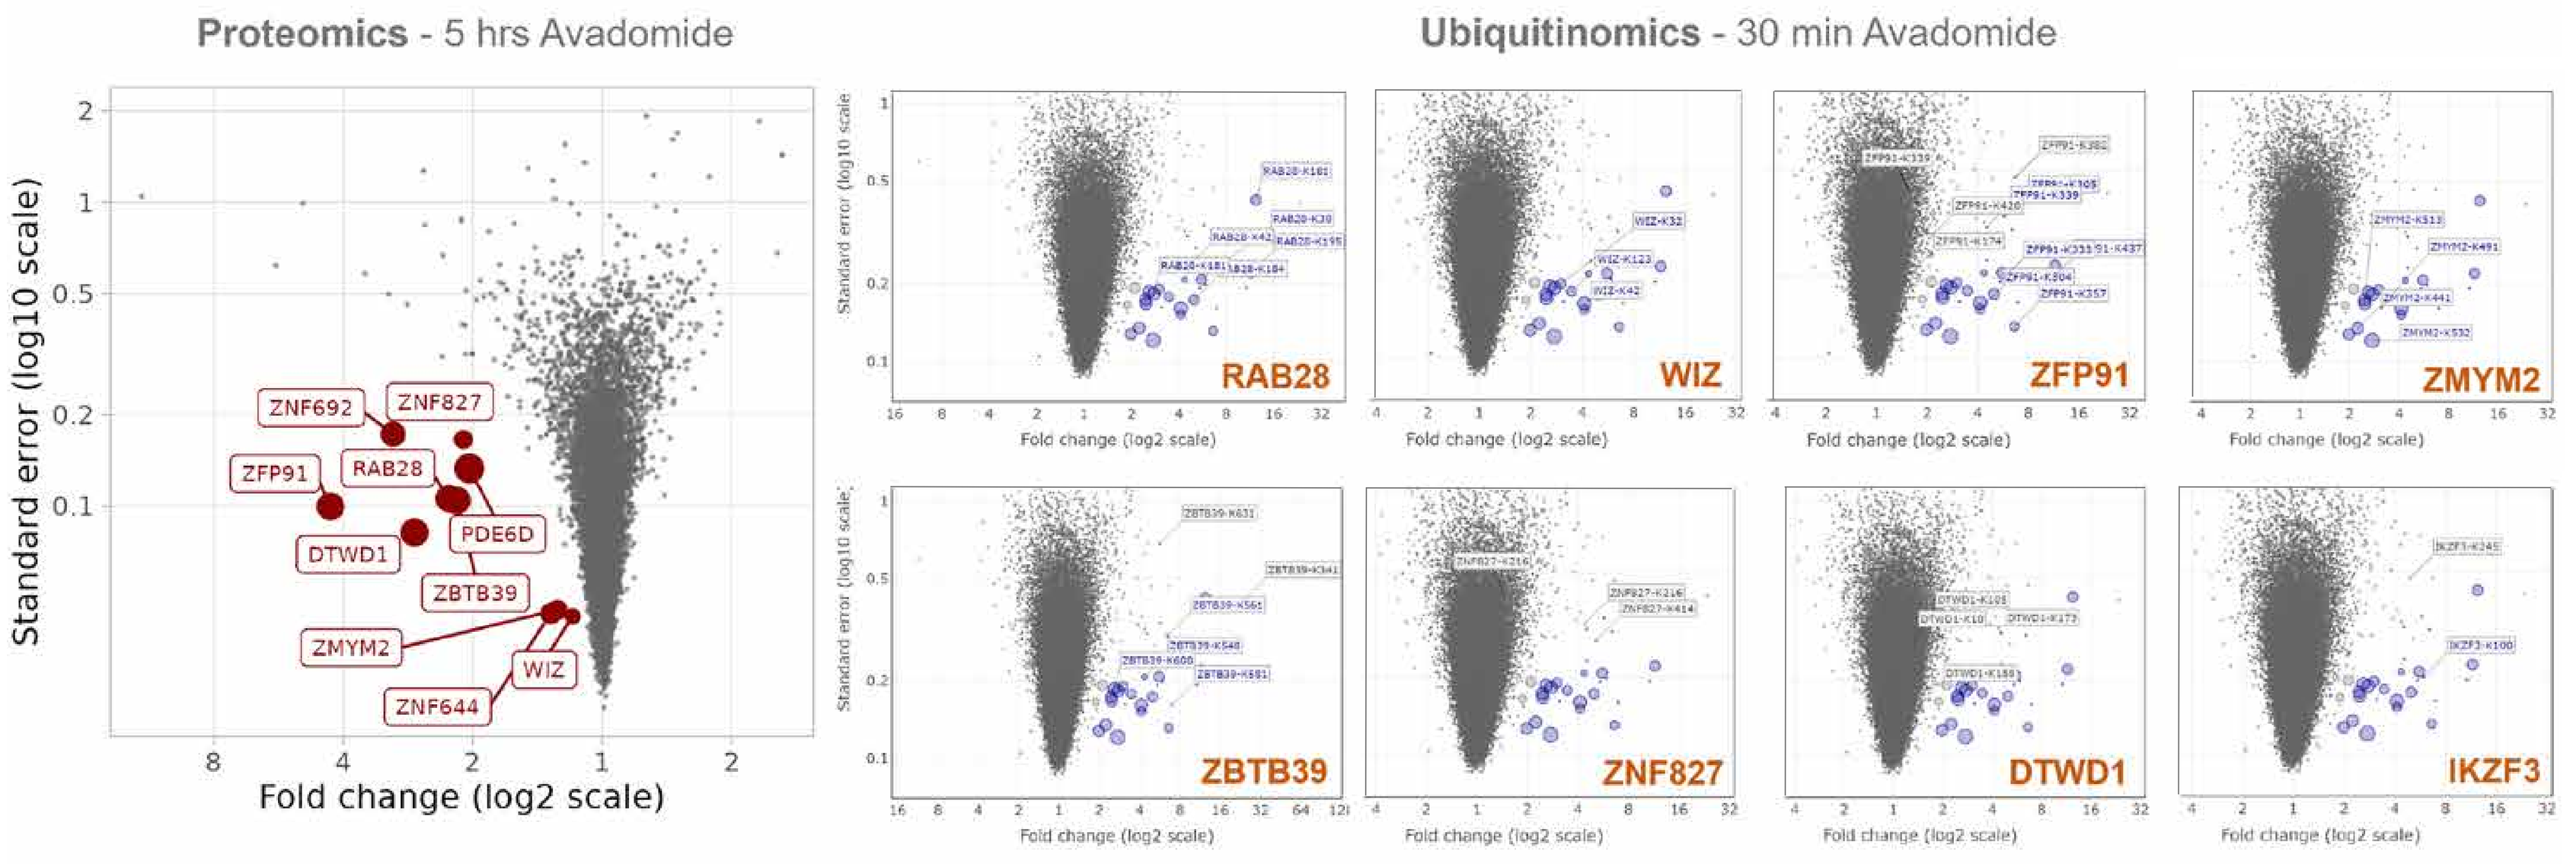
Proteomics - 5 hrs Avadomide
Ubiquitinomics - 30 min Avadomide
Standard error (log10 scale)
Fold change (log2 scale)
ZNF692
ZNF827
ZFP91
RAB28
DTWD1
PDE6D
ZBTB39
ZMYM2
WIZ
ZNF644
RAB28
WIZ
ZFP91
ZMYM2
ZBTB39
ZNF827
DTWD1
IKZF3

Combined analysis of proteomics and ubiquitinomics reveals and validates primary degrader targets

HEK293 cells were treated with the cereblon modulator avadomide. Ubiquitinomics was performed using MS-based K-GG remnant profiling, enabling the quantification of up to 50,000 ubiquitination sites in single DIA-MS runs without cellular proteasome inhibition[3]. Ubiquitinomics revealed induced ubiquitination sites for almost all neosubstrates found to be degraded upon 5 hrs treatment of HEK293 cells. These included sites on IKZF3, a protein not detected in the proteomics experiment due to its extremely low expression in HEK293 cells. Ubiquitinomics allows the rapid validation of cellular downregulations being due to E3 ligase-neosubstrate relationships, by analyzing degrader drugs in endogenous cellular systems without the need for pharmacological intervention or genetic modification.

References
1 Meier F. et al, 2020, Nature Methods
2 Steger et al, 2022, Proteomics
3 Steger et al, 2021, Nat. Commun.